San Antonio Breast Cancer Symposium® - December 5-9, 2023

RELATE 2: A real-world observational study assessing the feasibility, acceptability and perceived impacts of treatment-specific versions of the OWise digital health tracking app in patients receiving olaparib in early breast cancer or trastuzumab deruxtecan in metastatic breast cancer.

Sophie McGrath[1], Peter Hall[2], Michaël Acquadro[3], Soren Skovlund[3], Parveen Jayia[4], Stuart McIntosh[5], David Cameron[2]

1. The Royal Marsden Hospital, Surrey, UK; 2. Edinburgh Cancer Research Centre, Institute of Genetics and Cancer, University of Edinburgh, Edinburgh, UK; 3. Patient-Centered Research, Evidera; 4. AstraZeneca UK, London; 5. Belfast City Hospital, Belfast Health & Social Care Trust, School of Medicine, Dentistry and Biomedical Sciences, Queens University Belfast, Northern Ireland

Poster #: PO4-17-07

Plain language summary

Why are we performing this research?
- OWise is a digital health support and tracking app which has been shown to provide useful support in patients with breast cancer (BC)[1]
- OWise has been extended with two treatment-specific areas to support patients prescribed: trastuzumab deruxtecan (T-DXd) for human epidermal growth factor receptor 2 (HER2)-positive metastatic breast cancer (mBC)[2]; or olaparib for high risk early HER2-negative BC having mutations in the *BRCA1/2* genes[3]
- The current study assesses these digital support tools when used by patients with BC in routine clinical practice in the United Kingdom (UK)

How are we performing this research?
- RELATE 2 is a real-world observational research study
- Patients can sign up and participate in the study via the freely available OWise app and through interviews; healthcare professionals (HCPs) involved in BC care who participate in the study will also contribute data via interviews
- This study uniquely combines ongoing real-world data collection with the evaluation of a patient support app to meet patient-identified support needs relating to specific medical therapies in BC

Who will participate in this study?
- Patients receiving olaparib in early BC or T-DXd in mBC as part of routine clinical practice

Where can I access more information?
- This study is ongoing and no results are available
- More information about the RELATE 2 study is available by emailing: RELATE2@astrazeneca.com; more information about the OWise app is available from: info@owise.co.uk. You may also speak to your doctor about clinical studies

Background

- BC is the most common form of cancer in the UK, with around 56,000 new cases diagnosed each year[4]
- Effective use of medications for BC requires the education and active engagement of both patients and HCPs combined with ongoing vigilance for drug-related adverse reactions, which may negatively impact patient wellbeing and clinical outcomes
- OWise is a digital patient support app which can: 1) collect patient-reported and patient-generated data for real-world evidence (RWE) studies; and 2) support patients with a range of features, including treatment-specific information and guidance after prescription
- OWise has been clinically validated in patients with BC[1] and the app is approved by the UK's National Health Service (NHS) and is used as a case study example in the National Institute for Health and Care Excellence (NICE) Evidence Standards Framework for Digital Health Technologies[5]
- A previous qualitative study evaluating OWise in the Netherlands showed BC patients and healthcare providers found the tool usable and felt it had the potential to help patients take in more information from consultations, manage appointments, and feel more in control during treatment[1]
- In a systematic review of mobile apps available in Germany for supporting BC patients during treatment and aftercare, OWise received the highest mean score from a panel of three independent examiners from different institutions[6]
- Recently, two treatment-specific versions of the OWise app have been developed for supporting patients prescribed either: T-DXd for HER2-positive unresectable or mBC[2]; or olaparib for high risk early HER2-negative BC having germline mutations in the *BRCA1/2* genes[3]
 - It is hypothesized that OWise could positively impact these patients' confidence, self-monitoring of symptoms and experience of care while also generating RWE data that can assist HCPs and market authorisation holders to address gaps in patient care and education without increasing burden on the UK NHS

Rationale and study design

- RELATE 2 is a real-world observational research study to evaluate the impacts on patient experience, treatment use, and quality of care of two treatment-specific extensions of the OWise app in routine BC care in the UK (Figure 1)
- Eligible patients can sign up to one of the treatment-specific OWise versions using a leaflet obtained from their hospital providing details of the modules and an access code
- The study encourages patient engagement and is being conducted across two workstreams:
 - **Workstream 1** involves collection of participants' OWise fully anonymized data (including patient entered treatment data as well as usage statistics) and a brief survey for participants to provide feedback on their experience using OWise every three months for up to 18 months. Survey questions are focused on their perceptions of the usefulness of the app for monitoring their health changes, communicating with their care team, and their response to treatment. The surveys include two short patient-reported outcome (PRO) questionnaires, the EQ-5D 5L and the Patient's Global Impression of Treatment Tolerability (PGI-TT)
 - **Workstream 2** will launch approximately three months after the launch of Workstream 1 and will include qualitative interviews with two groups of people: 1) a small number of participants who are already enrolled in Workstream 1; and 2) clinicians (and multidisciplinary team/nurses) to further understand the perceived benefits of the app
- Participants will enter data into OWise actively about how they respond to their treatment, but also consent to the passive collection of data about how they use the OWise app
- All user data and survey responses are fully anonymous and are not linked to any personal identifiable details or NHS medical records, therefore only the patients are able to share their information recorded in the app with their HCPs
- All data collected in this study will be strictly confidential, in accordance with European data protection legislation, including country-level regulations and General Data Protection Regulation (GDPR)

Figure 1. Treatment-specific versions of the OWise digital healthcare app being used in RELATE 2 to support BC patients prescribed T-DXd (a) or olaparib (b)

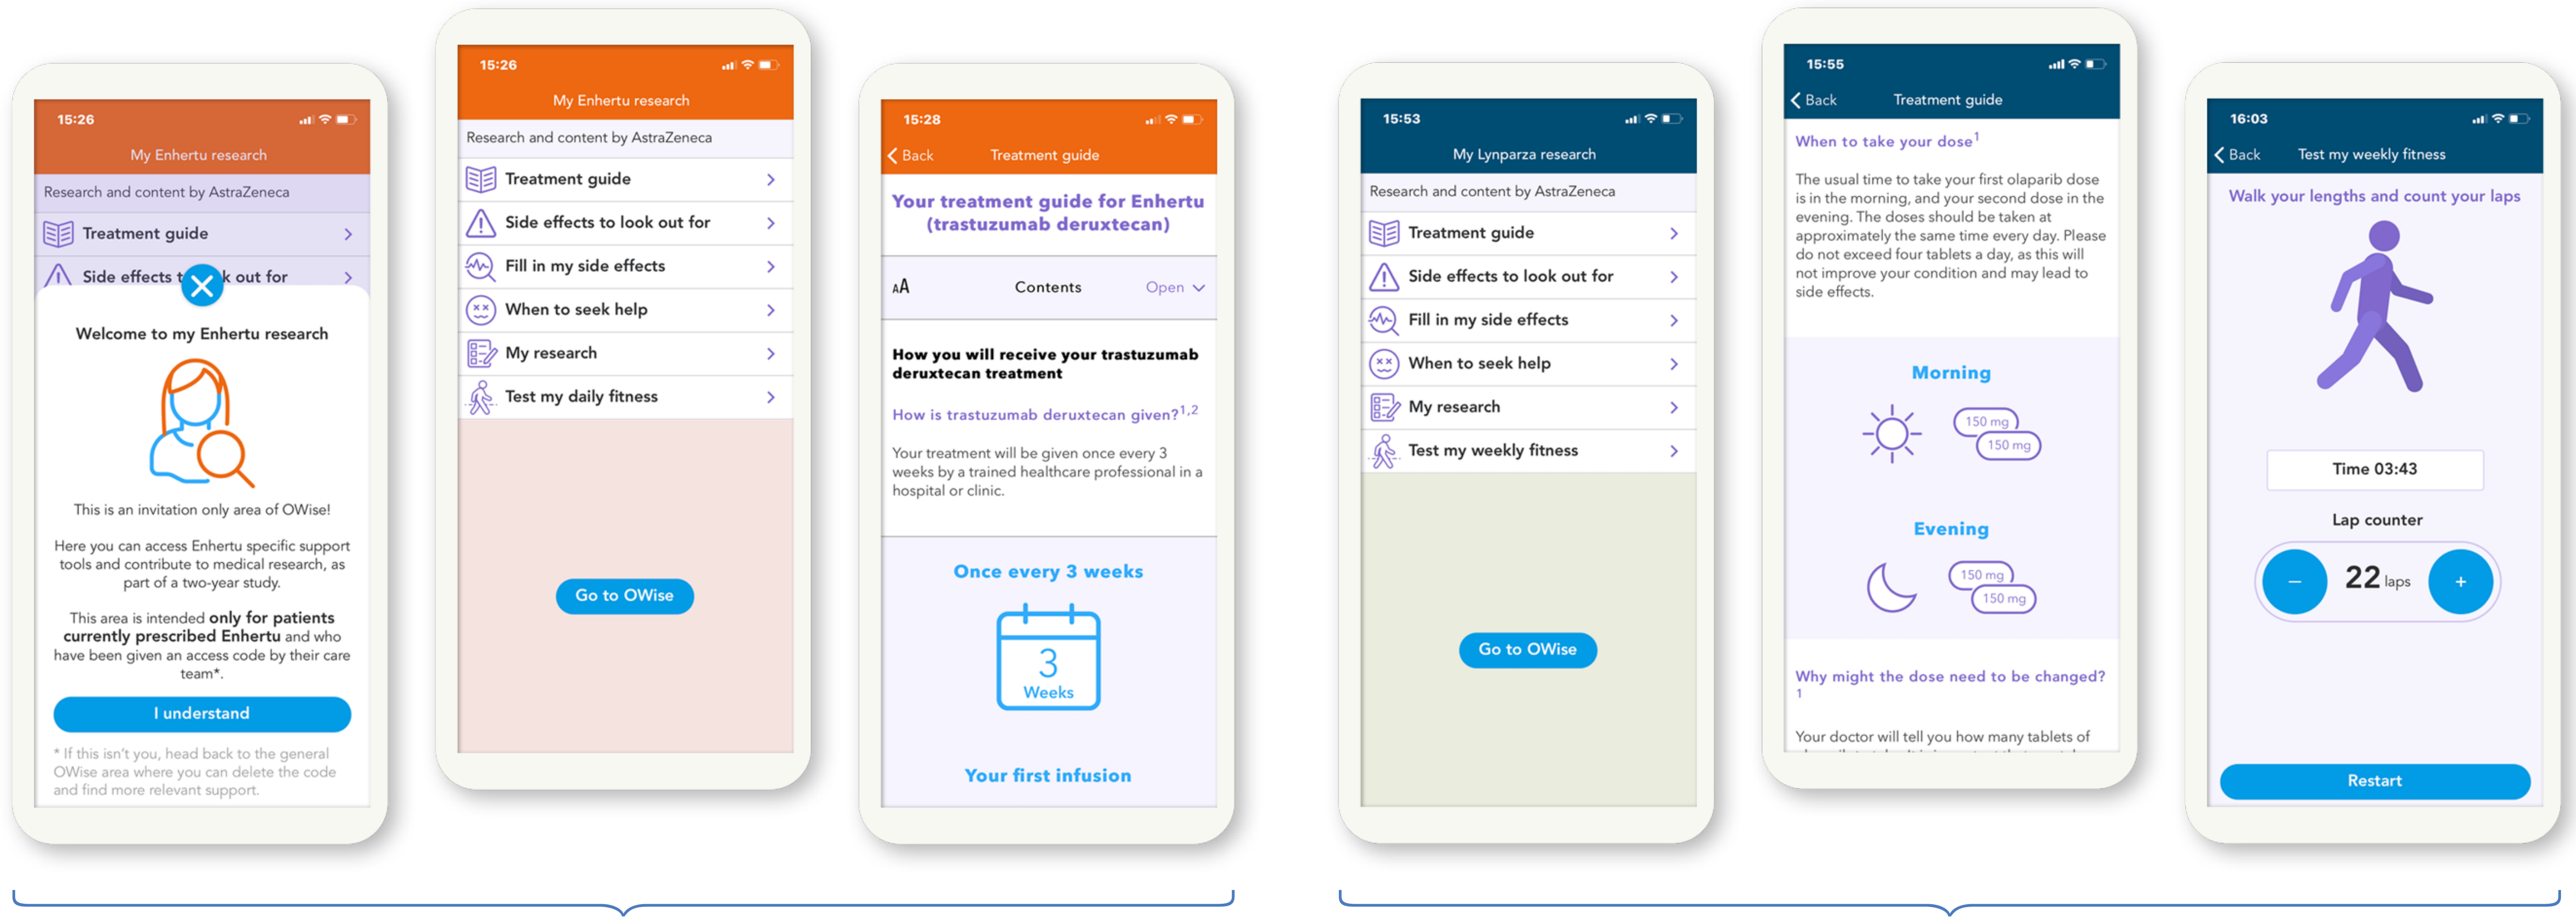
My Enhertu research
Research and content by AstraZeneca
Treatment guide
Side effects to look out for
Fill in my side effects
When to seek help
My research
Test my daily fitness
Welcome to my Enhertu research
This is an invitation only area of OWise!
Here you can access Enhertu specific support tools and contribute to medical research, as part of a two-year study.
This area is intended only for patients currently prescribed Enhertu and who have been given an access code by their care team*.
I understand
Go to OWise
Your treatment guide for Enhertu (trastuzumab deruxtecan)
How you will receive your trastuzumab deruxtecan treatment
How is trastuzumab deruxtecan given?
Once every 3 weeks
Your first infusion
My Lynparza research
Test my weekly fitness
When to take your dose
Morning
Evening
150 mg
Why might the dose need to be changed?
Walk your lengths and count your laps
Time 03:43
Lap counter
22 laps
Restart

a) Screenshots of T-DXd specific version of the OWise mobile app

b) Screenshots of olaparib specific version of the OWise mobile app

Study objectives

Primary objectives:
- Evaluate feasibility, reach, and uptake of treatment-specific versions of OWise in routine BC care in the UK
- Evaluate patient acceptability of treatment-specific versions of the OWise app
- Evaluate the perceived impact of OWise on aspects of patient-centred care, specifically use of patients' self-reported data by HCPs to improve care quality and safety
- Evaluate patient-reported experiences and impacts related to use of the treatment-specific OWise versions and explore associations between patient experiences and usage/usage patterns of OWise

Secondary objectives:
- Understand prescribing patterns for the treatments T-DXd and olaparib, and the use of concomitant medicine(s) in UK practice
- Describe demographic and clinical characteristics of patients treated in the UK real-world setting and using the OWise app
- Understand safety and tolerability profiles and strategies used in the real-world setting to manage adverse events among users of the OWise app

Key inclusion criteria for patients

- Be at least 18 years of age
- Have recently been treated with either T-DXd for HER2-positive unresectable or mBC, or olaparib for high risk early HER2-negative BC having germline mutations in the *BRCA1/2* genes
- Be able to create an OWise account (e.g., mobile or web application) and have activated the unique study code for the treatment specific version of OWise
- Be able to provide online informed consent for study participation

Statistical methods

- Participants will be invited to enter a survey via OWise at baseline and every three months (for up to 18 months)
- Aggregated anonymized participant-level quantitative data and usage data will be analyzed for response frequencies, means, standard deviations, medians, ranges, and confidence intervals
- Descriptive statistics and advanced statistics will be used to identify core variables and trends, investigate data distribution links between data, and test significant differences between datasets according to profile/status
- Interim data analyses will be conducted approximately 9 months following study launch, with final database lock at 21 months
- The target for accrual is 300 patients receiving T-DXd and 150 patients receiving olaparib to use the OWise app within this real-world study

Acknowledgements

This study is sponsored by AstraZeneca UK. We thank all participating patients and study team involved in the RELATE 2 real-world observational study. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for trastuzumab deruxtecan (T-DXd; DS-8201). In July 2017, AstraZeneca entered into a global development and commercialization collaboration with Merck for olaparib. Medical writing support in the preparation of this poster was provided by NexGen Healthcare Communications (London, UK) with financial support from AstraZeneca UK.

Conflicts of interest

Parveen Jayia is an employee of AstraZeneca UK

References

1. Young-Afat DA, et al. JMIR Cancer. 2016;2(1):e8.
2. Enhertu (trastuzumab deruxtecan). Summary of Product Characteristics.
3. Lynparza (olaparib). Summary of Product Characteristics.
4. Breast Cancer UK (2022). https://www.breastcanceruk.org.uk/about-breast-cancer/facts-figures-and-qas/facts-and-figures/. Accessed Nov 2023.
5. York Health Economics Consortium (2019). https://www.nice.org.uk/ Media/Default/About/what-we-do/our-programmes/evidence-standards-framework/evidence-case-studies.pdf. Accessed Nov 2023.
6. Gomm SIM, et al. Geburtshilfe Frauenheilkd. 2022;82(9):941-54.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster

This presentation is the intellectual property of the authors. Contact the presenter at sophie.mcgrath@rmh.nhs.uk for permission to reprint and/or distribute.